Shannon Entropy of Tau-PET Spreading Patterns Predict Alzheimer's Disease Progression in Amyloid-Positive APOE4 Carriers

Angélica Álvarez [1], María C. Ibañez [1], María Margarita Venegas [1], María Alejandra Tangarife [1], Paola Ruiz Puentes [1], Ram Mukunda [1].
[1] IGC Pharma

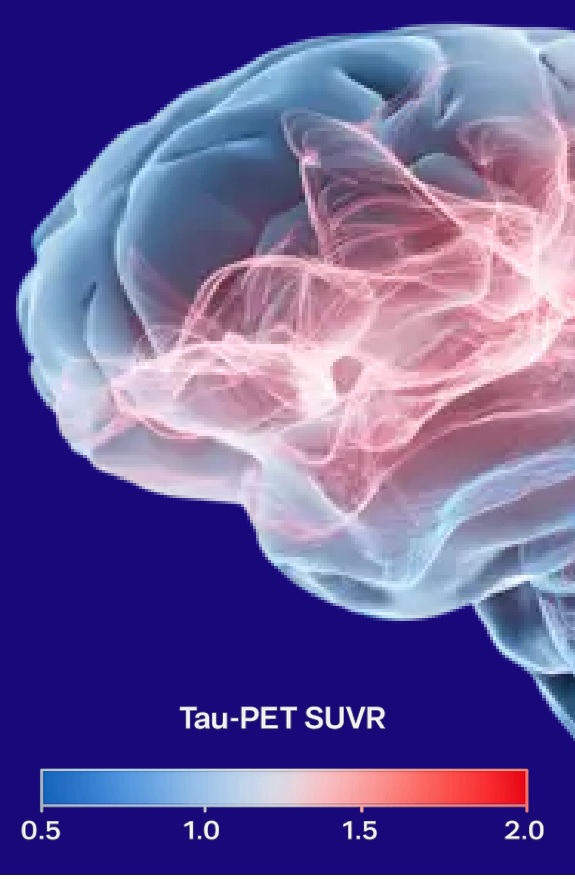
Tau-PET SUVR
0.5
1.0
1.5
2.0

1 Introduction

Tau-PET reveals substantial heterogeneity in tau distribution in Alzheimer's disease (AD). Discrete subtypes have been proposed, but a continuous and clinically interpretable metric is lacking.

We applied Shannon entropy to quantify tau dispersion across Braak regions and examined associations with cognition, longitudinal tau accumulation, amyloid status, and APOE genotype.

Braak Regions (bilateral averages)

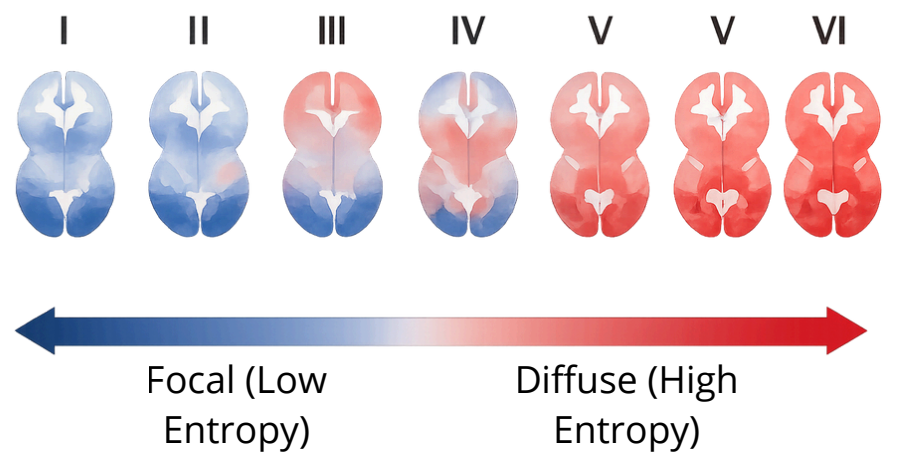
I
II
III
IV
V
V
VI
Focal (Low Entropy)
Diffuse (High Entropy)

Shannon Entropy (Normalized)

$$H = \frac{\sum_{i=1}^{6} p_i \log_2(p_i)}{\log_2(6)}$$

Range: [0, 1]
Lower = focal
Higher = diffuse

2 Methodology

931 ADNI participants (1,570 scans) ^{18}F-AV-1451 tau-PET (UC Berkeley pipeline)

Six bilateral Braak-stage regions (I–VI) SUVR composites

Shannon entropy of regional distribution normalized by $\log_2(6)$ → [0,1]

Temporally matched to tau-PET (±180 days; n=869)

A+ (amyloid positive): CSF Aβ42 <976 pg/mL (n=403) T+ (high tau): upper tertile of composite SUVR (>1.22)

A+T+ (amyloid-positive, tau-positive): n=128. Entropy dichotomized at median within A+T+ Longitudinal A+ with follow-up ≥1.5 y: n=112 (mean follow-up 3.07±1.03 years) A–T+ (amyloid-negative, tau-positive): n=21

Analytical Workflow

Step 1
Construct 6 Braak-stage composites by bilateral averaging of FreeSurferdefined regional SUVRs

Step 2
Compute normalized Shannon entropy (continuous)

Step 3
Dichotomize entropy at median within A+T+

Step 4
Assess cross-sectional cognition and longitudinal tau accumulation

3 Results

FOCAL (LOW ENTROPY) PATTERNS ARE ASSOCIATED WITH WORSE COGNITION DESPITE SIMILAR TOTAL TAU BURDEN (A+T+ PARTICIPANTS)

Cognition by Entropy Group (A+T+)

	Focal (Low Entropy) n=64	Diffuse (High Entropy) n=64
MMSE (higher is better)	24.8 ± 3.6	26.8 ± 3.4
	p = 0.0004	
CDR-SB (higher is worse)	3.21 ± 2.6	1.69 ± 2.6
	p < -0.00001	

COMPOSITE SUVN

Total Tau Burden Focal (Low (Composite SUVR)

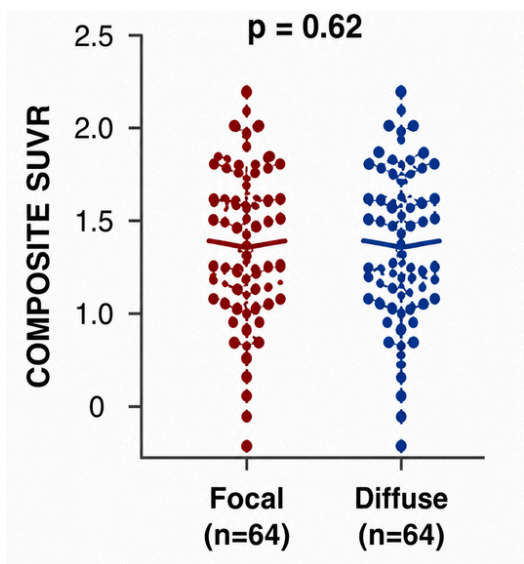
p = 0.62
COMPOSITE SUVR
2.5
2.0
1.5
1.0
0
Focal (n=64)
Diffuse (n=64)

Entropy Correlates with Cognition (A+T+)

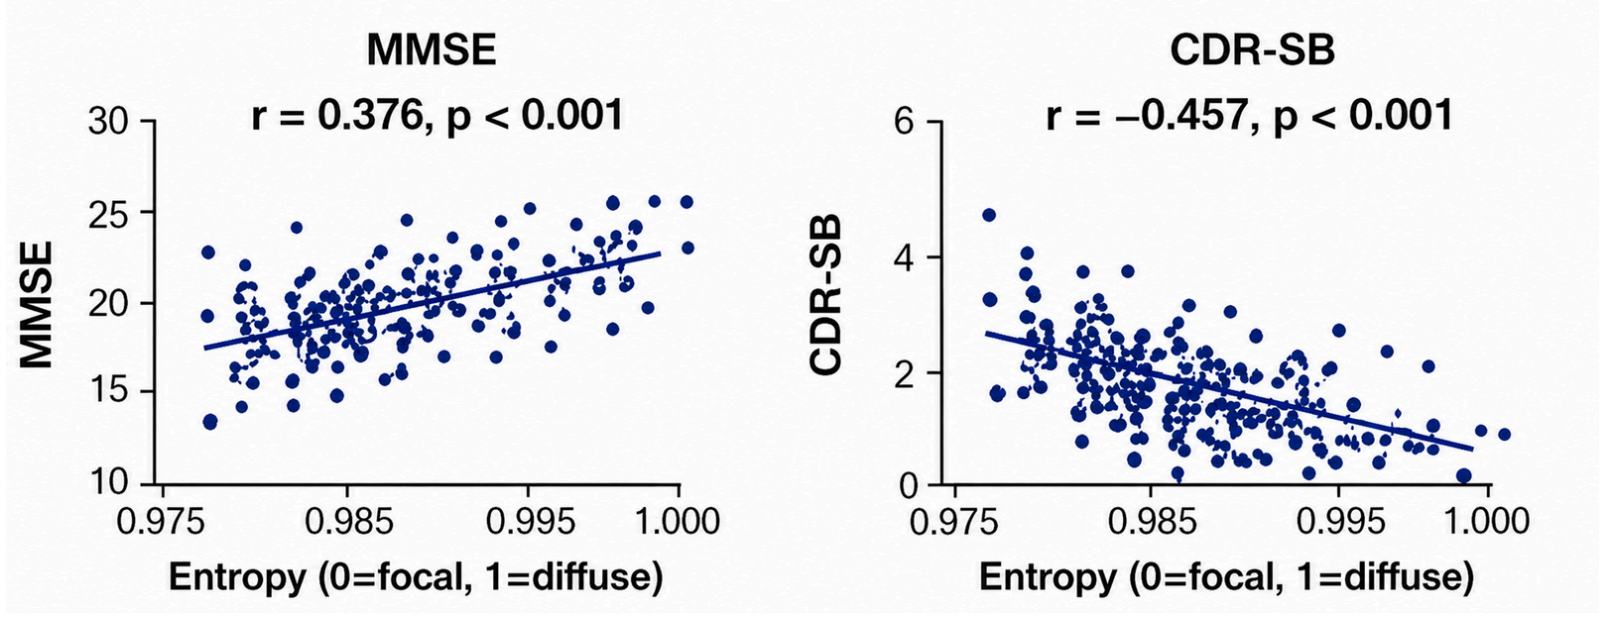
MMSE
r = 0.376, p < 0.001
MMSE
30
25
20
15
10
0.975
0.985
0.995
1.000
Entropy (0=focal, 1=diffuse)
CDR-SB
r = –0.457, p < 0.001
CDR-SB
6
4
2
0
0.975
0.985
0.995
1.000
Entropy (0=focal, 1=diffuse)

COGNITIVE IMPACT OF ENTROPY IS APOE4-DEPENDENT (PRIMARY RESULT)

CDR-SB by Entropy Group Within Genotype (A+T+)

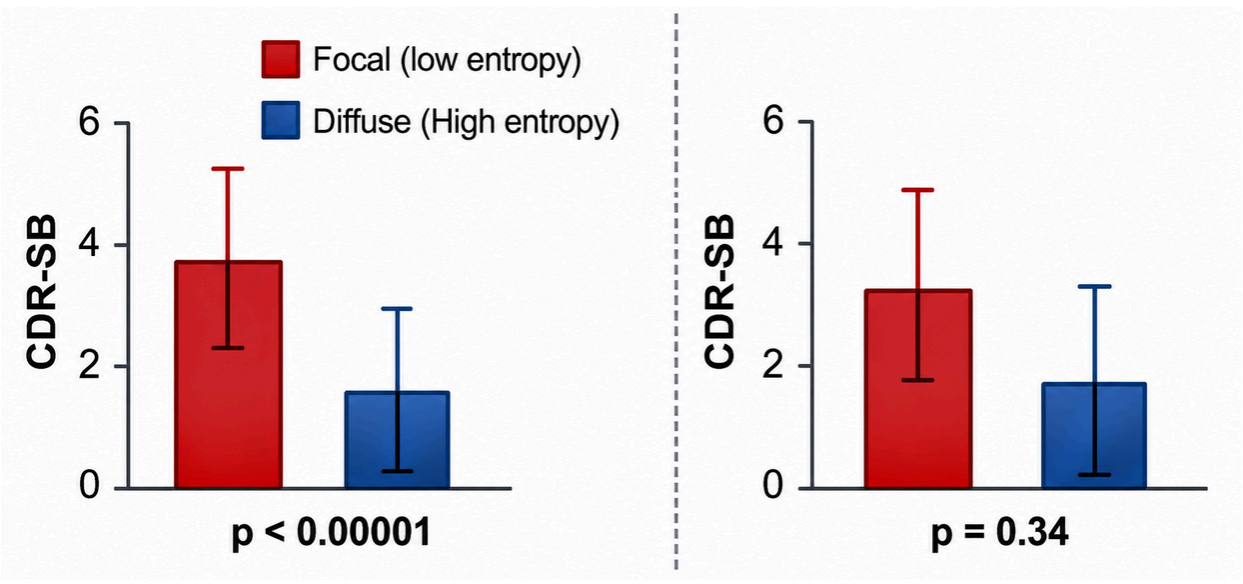
Focal (low entropy)
Diffuse (High entropy)
CDR-SB
6
4
2
0
p < 0.00001
CDR-SB
6
4
2
0
p = 0.34

★ **Focal vs. diffuse differences in cognition occur despite similar total tau burden, and this effect is present only in APOE4 carriers.**

4 Results (Longitudinal)

LOWER BASELINE ENTROPY PREDICTS FASTER TAU ACCUMULATION IN AMYLOID-POSITIVE PARTICIPANTS

Correlation: Baseline Entropy vs. Annual Tau Accumulation (A+)

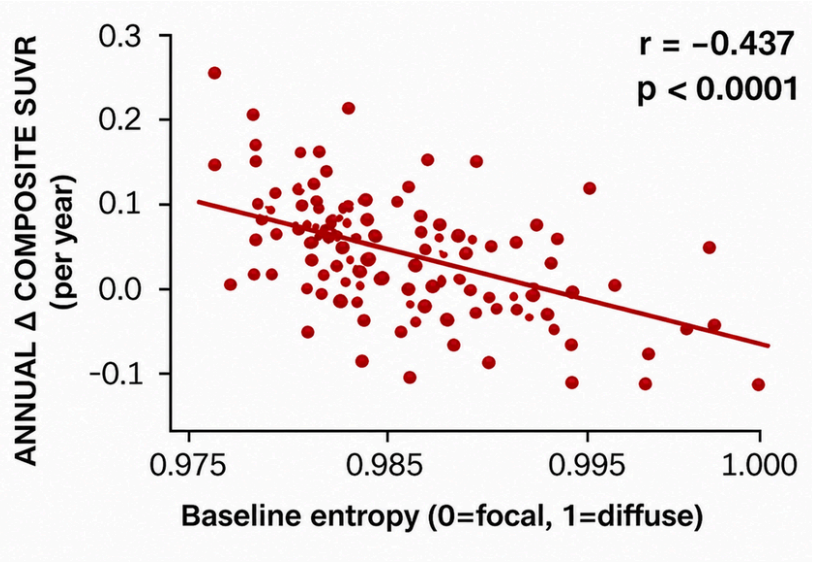
r = –0.437
p < 0.0001
ANNUAL Δ COMPOSITE SUVR (per year)
0.3
0.2
0.1
0.0
-0.1
0.975
0.985
0.995
1.000
Baseline entropy (0=focal, 1=diffuse)

Annual Tau Accumulation by Entropy Group (A+)

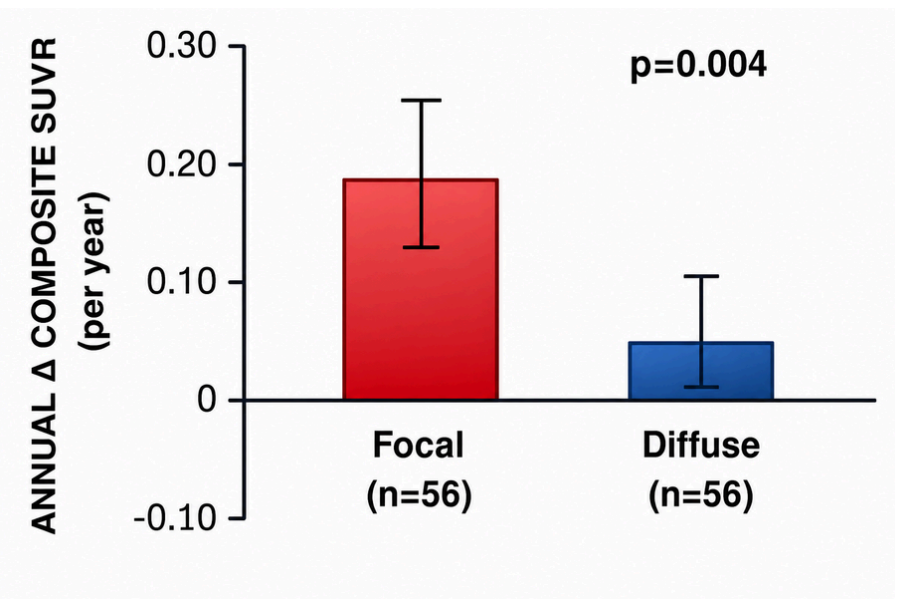
p=0.004
ANNUAL Δ COMPOSITE SUVR (per year)
0.30
0.20
0.10
0
-0.10
Focal (n=56)
Diffuse (n=56)

★ **Group-level:** Focal patterns show significantly greater tau accumulation than diffuse patterns in amyloid-positive participants.

No association in amyloid-negative participants
r = 0.06, p = 0.82

5 Results (Amyloid-Negative, Tau-Positive)

A–T+ Participants (n=21)

Preserved Cognition
MMSE and CDR-SB within normal to mildly impaired ranges

Lower APOE4 frequency
28.6% in A–T+ vs. 71.9% in A+T+ p = 0.0003

6 Conclusions

- Shannon entropy may provide a concise and robust measure of tau-PET spatial heterogeneity.
- Focal tau patterns could identify amyloid-positive APOE4 carriers with greater clinical impairment and faster tau accumulation—despite similar total tau burden.
- Preserved cognition in A–T+ participants and lower APOE4 frequency support the amyloid-dependent clinical impact of tau.
- Entropy may improve patient stratification for disease monitoring and therapeutic trials.

7 References

- Shannon CE. Bell Syst Tech J. 1948;27:379–423.
- Braak H, Braak E. Acta Neuropathol. 1991;82:239–259.
- Vogel JW, et al. Nat Med. 2021;27:871–881.
- Jack CR Jr, et al. Alzheimers Dement. 2024;20:5143–5169. Ossenkoppele R, et al. JAMA Neurol. 2021;78(8):961–971.

★ Entropy could capture clinically meaningful heterogeneity in tau spread. In amyloid-positive APOE4 carriers, focal tau patterns may forecast worse outcomes—earlier.